Effect of rumen-protected carbohydrate (RUPCA) supplementation on blood and plasma metabolites in feedlot finishing steers during heat stress

Juan P. Russi*[1,3], Elias Peruzzo[1] , Nicolas DiLorenzo[2] , and Alejandro E. Relling[1] ,

[1] Facultad de Cs Veterinarias, UNLP, Buenos Aires, Argentina, [2] University of Florida, NFREC, Marianna, FL,
[3] RUPCA LLC, Merced, CA

ASAS Abstract #: M431 – Orlando, July 12-16, 2015

INTRODUCTION

Finishing steers during the summer can be challenging. The effect of high temperatures and humidity, solar radiation and wind speed dramatically affect DMI, ADG and ultimately animal performance (Mader et al., 2010) . Independently of diet constituents, when the animal suffers from heat stress not only the DMI decreases but it is also metabolically challenged altering blood and plasma metabolites (Rhoads et al., 2009).

The objective of this study was to evaluate the inclusion of a rumen protected carbohydrate (RUPCA) (US Patent # 8,507,025) on blood and plasma metabolites of finishing steers during heat stress.

MATERIALS AND METHODS

One hundred and thirty five crossbred steers, initial body weight (BW) 287±13 kg were used in a 62-d experiment. Temperature humidity index (THI) and cattle panting score (0-4) were measured every day during the experiment and averaged 72 ± 4.9 and 1.15 respectively. All the animals were fed 91.4% of a basal diet (% DM), containing 22.3% corn silage, 65.9% dry corn, 0.6% sunflower meal, 0.5% urea, 2% minerals and vitamins. Steers were assigned to one of three treatments, T0) fed basal diet plus 8.6% of a supplement, T1) fed the basal diet plus 4.3% supplement and 4.3 % RUPCA and T2) fed basal diet plus 8.6 % RUPCA. The supplement and RUPCA contained (% DM) 58.1% soybean meal, 38.9% dextrose, 2% urea and 1% minerals salts, but they differed on the processing of the carbohydrate (i.e., protected or not from ruminal degradation). Blood samples were taken from 4 animals in each pen. Animals were sampled from the jugular vein prior to morning feeding and collected in tubes containing EDTA on d 0, 15, 39, and 62. Samples were maintained in a cooler until collection was finished. Glucose from blood was analyzed in situ with a glucometer (Optimum Xceedt, ABBOTT Lab Argentina). The plasma insulin concentration was analyzed with a RIA as described previously. The minimum detectable concentration was 0.05 ng/mL (Díaz-Torga, 2001). Plasma NEFA concentration was measured on d 0, 39 and 62, the protocol used was as described by Randox labs (FA 115 Randox Laboratories Ltd.). The minimum detectable concentration was 72 m*M*.

Coefficients of intra and inter samples were 7.48% and 23%. The urea plasma concentrations was measured on d 0, 39 and 62, the protocol used was as described by Wiener Lab (2R UREA Color). The minimum detectable concentration of urea was 0.02 g/L. Coefficients of intra and inter samples were 9.7% and 11%, respectively.

Insulin sensitivity was calculated for days 0, 39 and 62 using a Revised Quantitative Insulin Sensitivity Check Index (RQUICKI), which is calculated based on the blood plasma concentrations of glucose (Gb) in mg/dL, insulin (Ib) in µU/mL and free NEFA (NEFA) in m*M*, b denotes basal values. Formula RQUICKI = 1/[log (Gb) + log (Ib) + log (NEFA)] (Holtenius, 2007).

RESULTS

Table 1: Effect of rumen protected carbohydrate on blood glucose concentration, plasma insulin, NEFA and urea concentrations and Revised Quantitative Insulin Sensitivity Check Index (RQUICKI) RQUICKI on finishing steers during heat stress.

						P-Values	
Item	T0	T1	T2	SEM	Trt	day	Trt*day
Glucose, mg/dL	88a	83.b	83b	2.4	0.03	<0.0001	0.35
Insulin, ng/mL	0.62	0.68	0.68	0.056	0.69	0.0014	0.01
NEFA, m*M*	184	191	161	17.2	0.42	<0.0001	0.5
Urea, mg/dL	24	27	22	0.1	0.15	<0.0001	0.02
RQUICKI	0.301	0.305	0.307	0.005	0.945	<0.0009	0.04

Treatment*d interactions were found for insulin (P=0.01) urea (P=0.02) and RQUICKI. T0 showed higher blood glucose concentration (P=0.05).

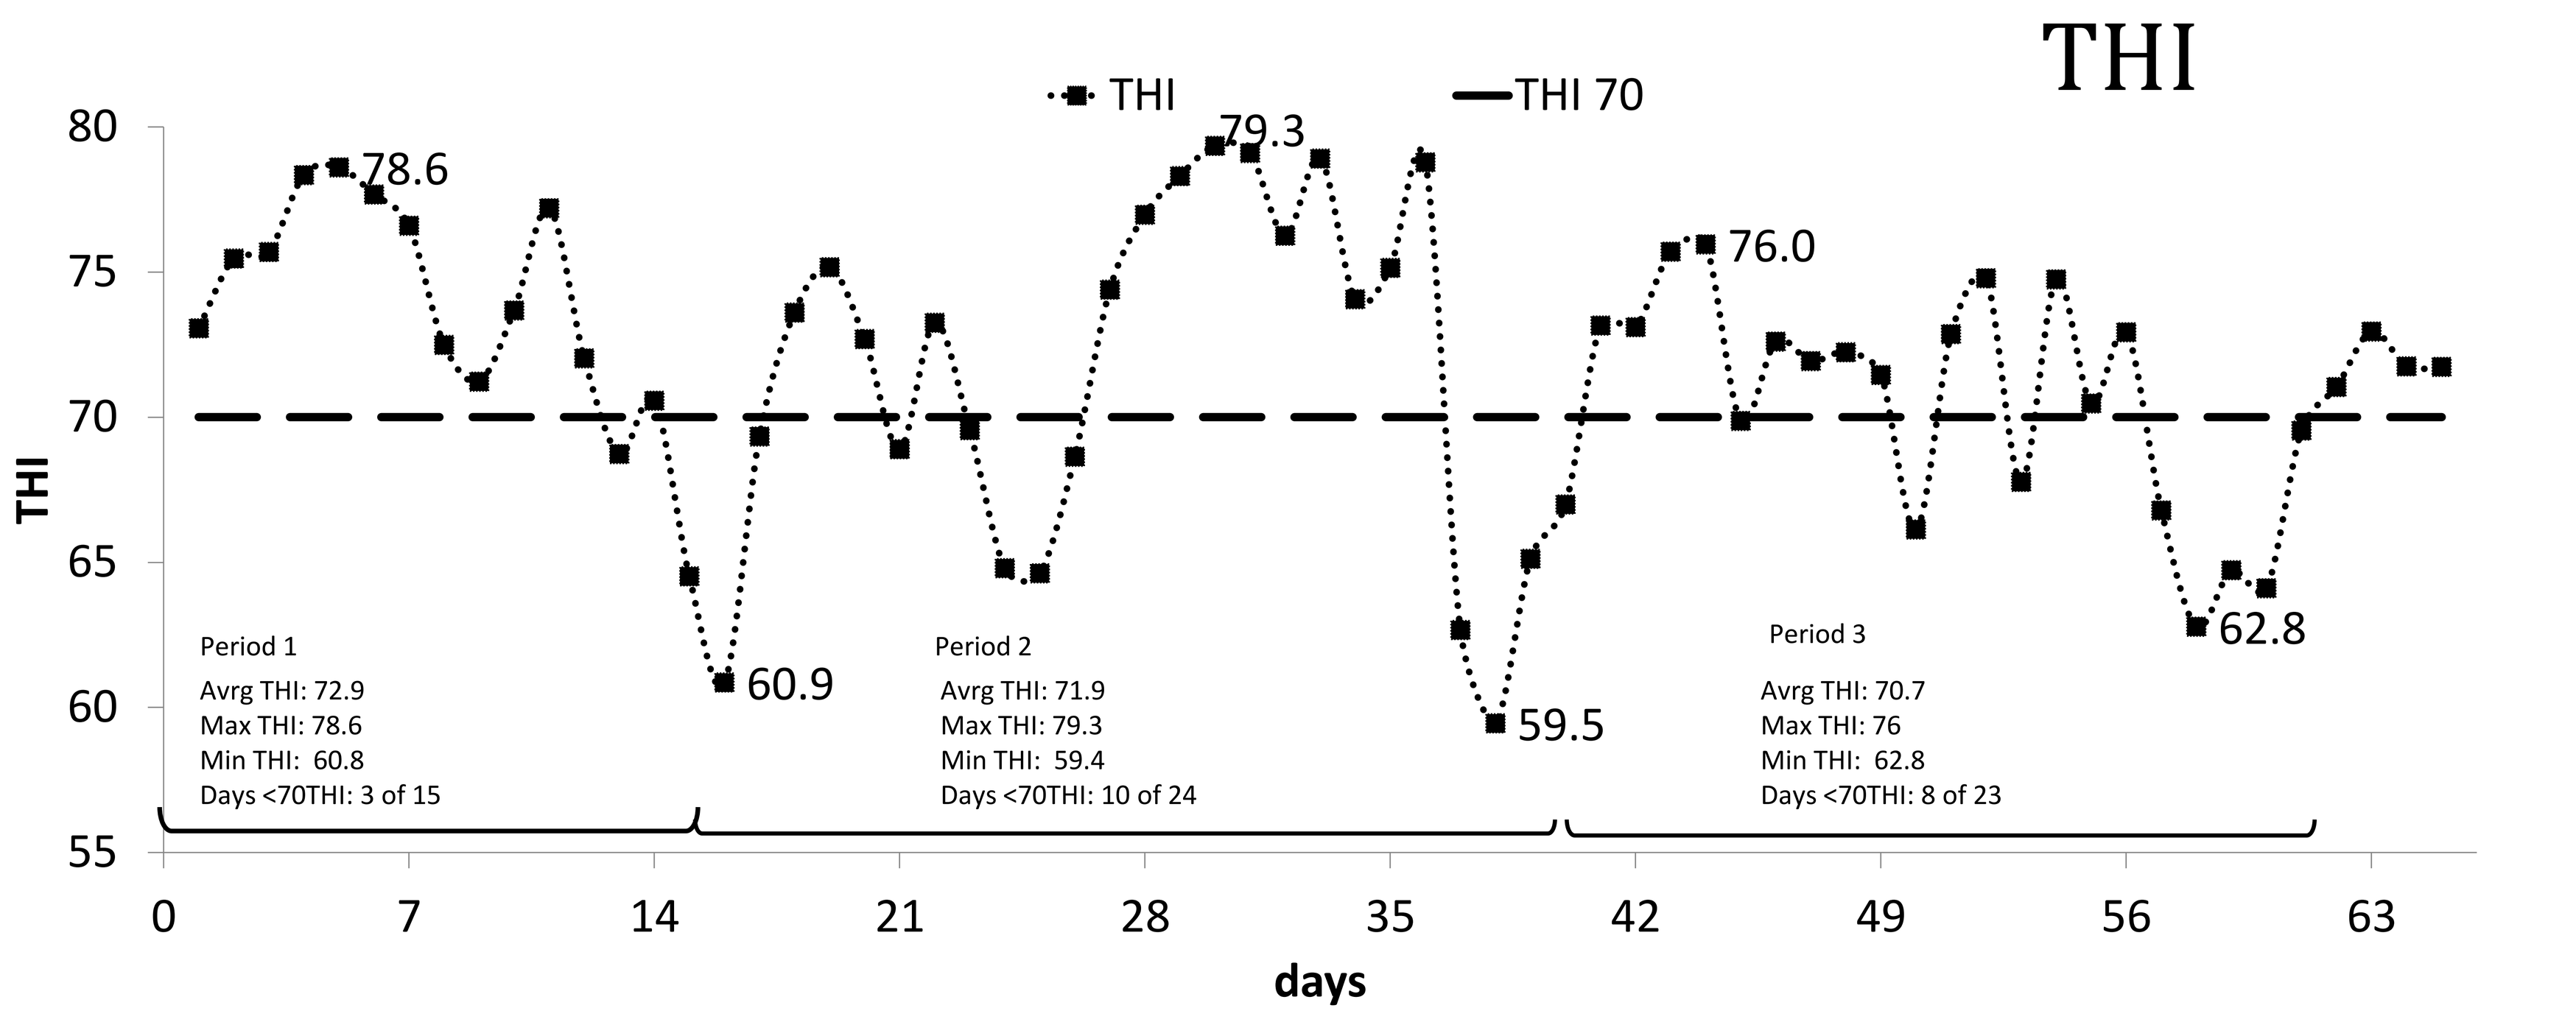
THI
THI
THI 70
80
75
70
65
60
55
78.6
79.3
76.0
60.9
59.5
62.8
Period 1
Avrg THI: 72.9
Max THI: 78.6
Min THI: 60.8
Days <70THI: 3 of 15
Period 2
Avrg THI: 71.9
Max THI: 79.3
Min THI: 59.4
Days <70THI: 10 of 24
Period 3
Avrg THI: 70.7
Max THI: 76
Min THI: 62.8
Days <70THI: 8 of 23
0 7 14 21 28 35 42 49 56 63
days

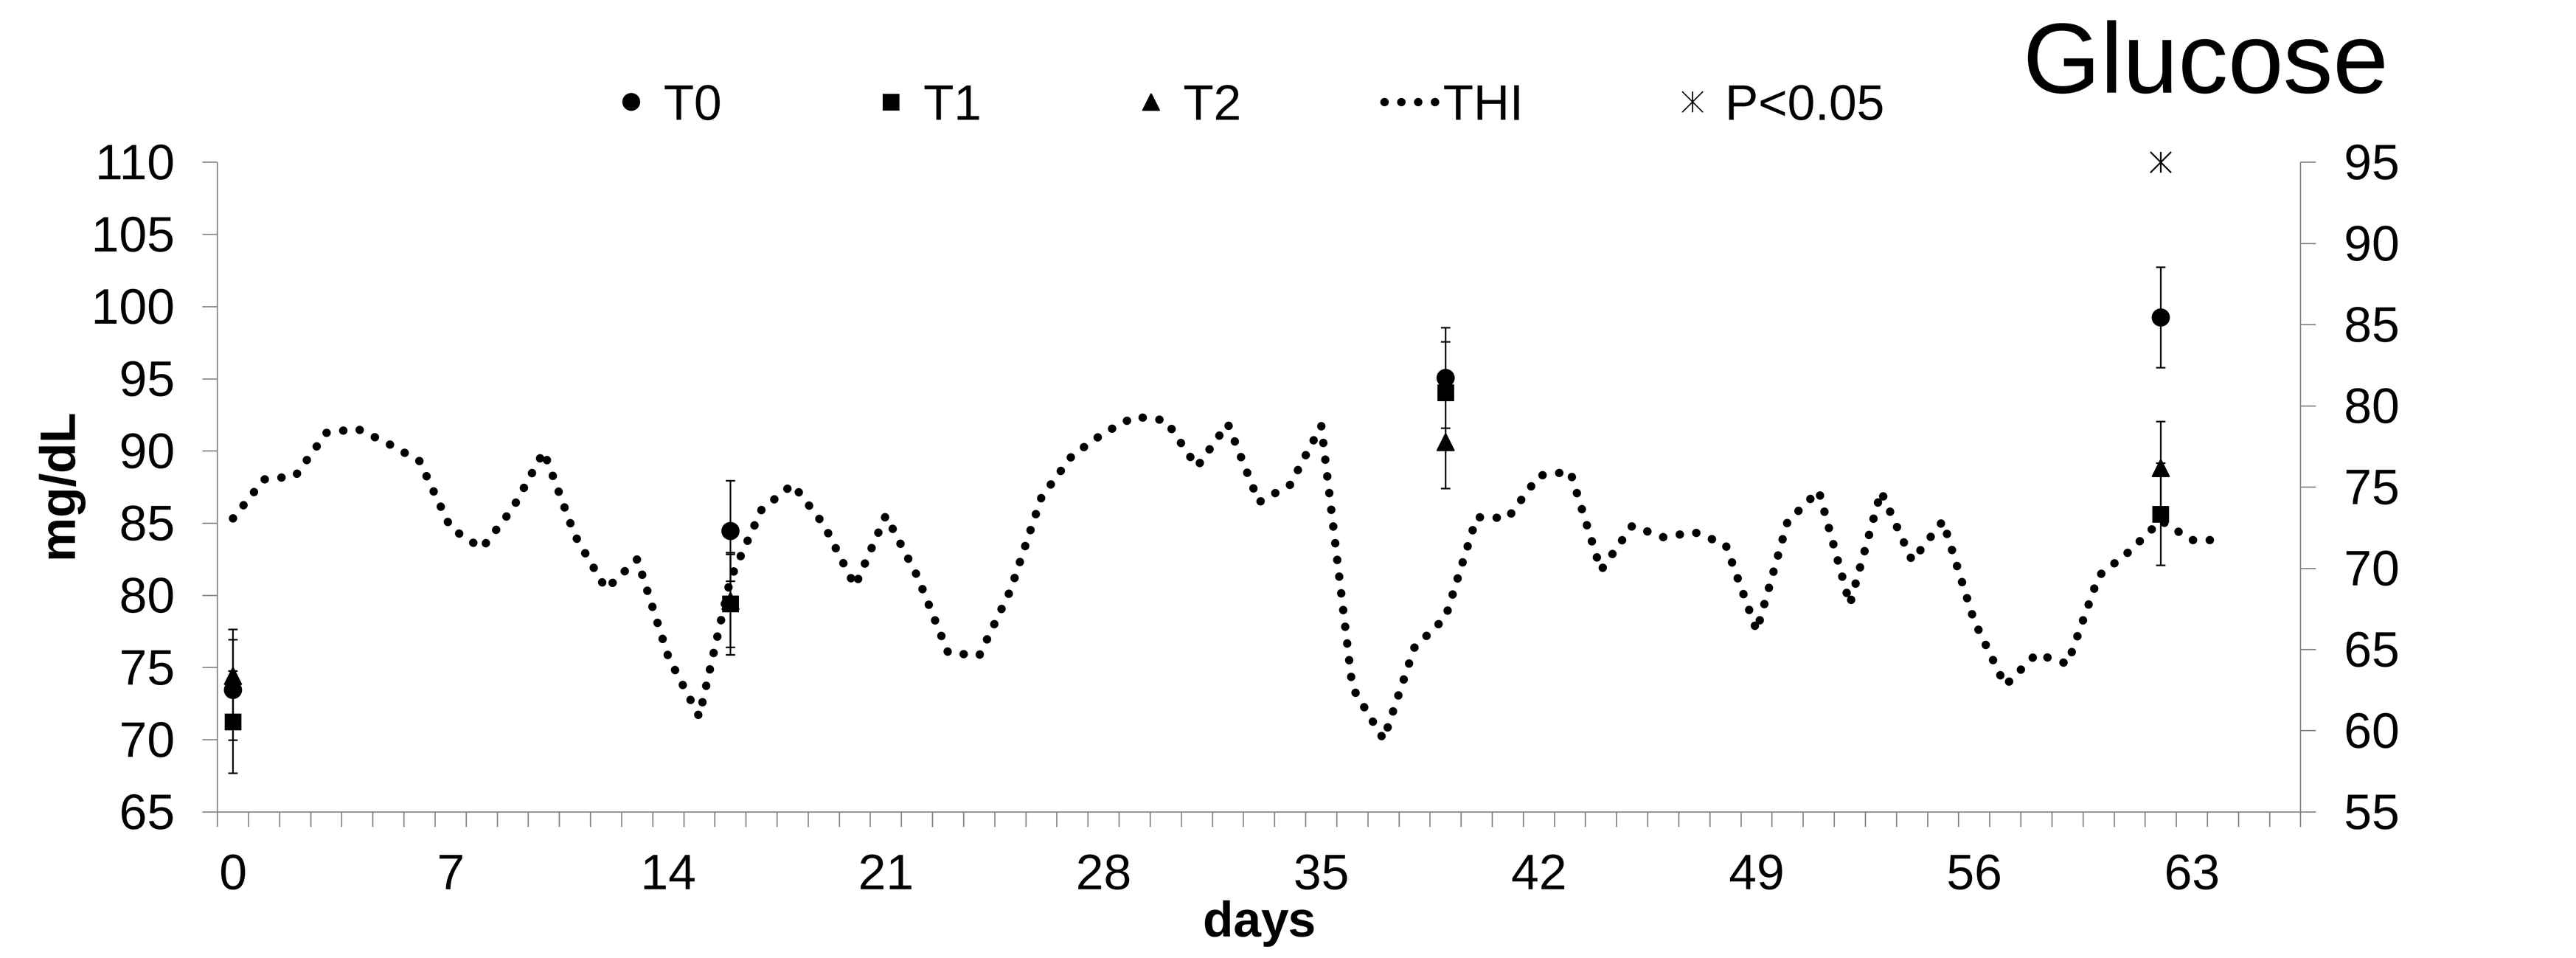
Glucose
T0
T1
T2
THI
P<0.05
mg/dL
110 105 100 95 90 85 80 75 70 65
95 90 85 80 75 70 65 60 55
0 7 14 21 28 35 42 49 56 63
days

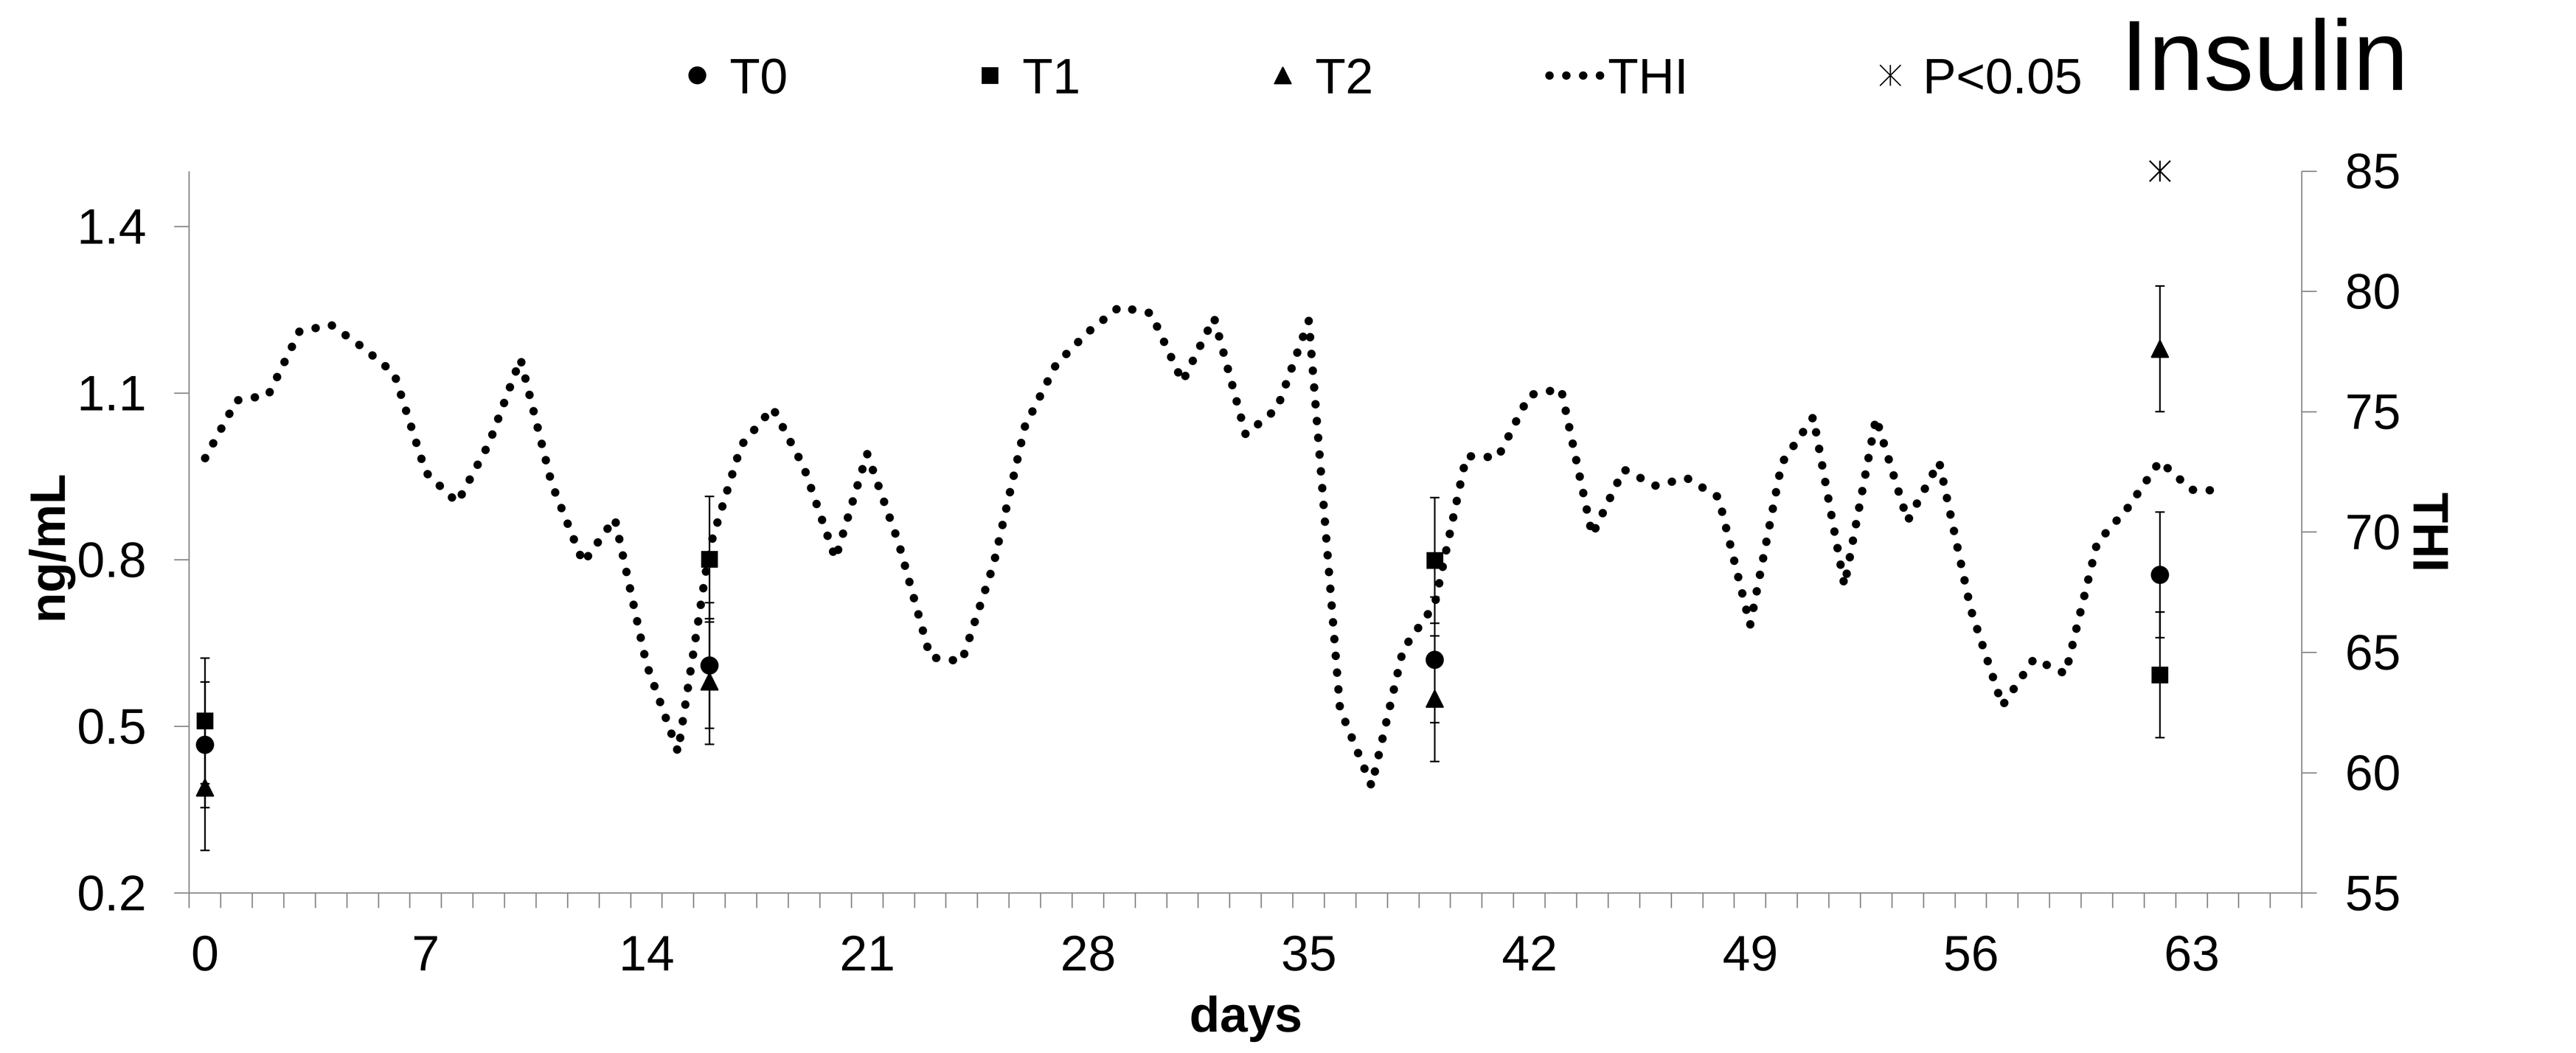
Insulin
T0
T1
T2
THI
P<0.05
ng/mL
1.4 1.1 0.8 0.5 0.2
85 80 75 70 65 60 55
THI
0 7 14 21 28 35 42 49 56 63
days

CONCLUSION

The results suggest that including RUPCA might help to mitigate the negative effects of heat stress, potentially improving animal performance.

REFERENCES

Mader, T. L. 2010 J. Anim. Sci. 88(6):2153-2165.
Rhoads, R. P. 2009. J. Anim. Sci. 91(6):2492-2503.
Díaz-Torga G.S. 2001. Theriogenology, 56((1)):111-122.
Holtenius, P. 2007. Acta Vet. Scand. 49(29).